8579

Efficacy and Safety of the Hedgehog Pathway Inhibitor Vismodegib in Patients With Advanced Basal Cell Carcinoma (BCC): ERIVANCE BCC Study Update

Aleksandar Sekulic,[1] Michael R Migden,[2] Anthony E Oro,[3] Karl Lewis,[4] John D Hainsworth,[5] Simon Yoo,[6] Luc Dirix,[7] Jeannie Hou,[8] Huibin Yue,[8] Axel Hauschild,[9] on behalf of the ERIVANCE BCC study investigators

[1]Mayo Clinic, Scottsdale, AZ, USA; [2]MD Anderson Cancer Center, Houston, TX, USA; [3]Stanford University School of Medicine, Stanford, CA, USA; [4]University of Colorado Cancer Center, Denver, CO, USA; [5]Sarah Cannon Research Institute, Nashville, TN, USA; [6]Northwestern University, Evanston, IL, USA; [7]Sint-Augustinus Hospital, Antwerp, Belgium; [8]Genentech, Inc., South San Francisco, CA, USA; [9]Universitätsklinikum Schleswig-Holstein, Kiel, Germany

ABSTRACT

Background: The ERIVANCE BCC study is the pivotal trial of vismodegib (GDC-0449), a first-in-class small-molecule inhibitor of Hedgehog signaling, for treatment of locally advanced (laBCC) and metastatic BCC (mBCC), for which there are no other effective therapy options. The study met the primary endpoint of overall response rate by independent review (Sekulic, Melanoma Res 2011).

Here we report a 6-mo update of investigator-assessed (I-A) efficacy and safety endpoints as of May 26, 2011.

Methods: This multicenter, international, nonrandomized 2-cohort study enrolled patients (pts) with laBCC (deemed inoperable or for whom surgery would be significantly disfiguring), and mBCC pts with RECIST-measurable disease. Pts received 150 mg oral vismodegib daily.

Results: 104 pts (71 laBCC/33 mBCC) enrolled at 31 sites in the US, Europe, and Australia. I-A efficacy endpoints were reported as follows.

	Nov 26, 2010 data cut-off		May 26, 2011 data cut-off	
Parameter	mBCC (n=33)	laBCC (n=63)	mBCC (n=33)	laBCC (n=63)
Overall response rate (ORR), n (%) (95% CI)	15 (45.5) (28.1–62.2)	38 (60.3) (47.2–71.7)	16 (48.5) (30.8–66.2)	38 (60.3) (47.2–71.7)
Median duration of response (DOR), mo (95% CI)	(n=15) 12.9 (5.55–12.91)	(n=38) 7.6 (7.43–NE)	(n=16) 12.9 (5.55–NE)	(n=38) NE (7.62–NE)
Median progression-free survival (PFS), mo (95% CI)	9.2 (7.39–NE)	11.3 (9.46–16.82)	9.3 (7.39–16.59)	12.9 (10.22–NE)

One-year survival rate was 77.3% (95% CI 62.48–92.09%) for mBCC and 93.1% (95% CI 86.49–99.63%) for laBCC. Adverse events (AEs) in >30% of pts were muscle spasms, alopecia, dysgeusia, weight decrease, fatigue, nausea, and amenorrhea (33.3%; 2/6 pts). Serious AEs were reported in 32 pts (31%). No additional fatal AEs were reported since the prior data cut-off (n=7, 7%; none considered related to vismodegib).

Conclusions: This 6-mo update of I-A efficacy and safety endpoints from the ERIVANCE BCC study supports the significant clinical benefit of vismodegib in both laBCC and mBCC reported at the primary analysis. Median DOR and PFS increased numerically with follow-up. The AE profile was consistent with the prior data cut. These results further support the efficacy of vismodegib for treatment of advanced BCC.

INTRODUCTION

- Basal cell carcinoma (BCC) is the most common human malignancy,[1] and while most BCCs are surgically managed, no effective therapy exists for metastatic BCC (mBCC) or locally advanced BCC (laBCC).
- Inappropriate activation of the Hedgehog signaling pathway plays a key role in the pathogenesis and progression of virtually all BCCs[2,3] and represents a novel therapeutic target for the treatment of mBCC or laBCC.[4]
- Vismodegib — a first-in-class small-molecule hedgehog pathway inhibitor — has been approved by the US Food and Drug Administration for the treatment of adults with mBCC, or with laBCC that has recurred following surgery or who are not candidates for surgery, and who are not candidates for radiation.
- The Phase II, pivotal study of vismodegib for the treatment of advanced BCC — ERIVANCE BCC — met its primary endpoint, with independently assessed objective response rates (ORRs) of 30% in patients with mBCC and 43% in patients with laBCC.[5]

OBJECTIVE

- The primary analysis of the ERIVANCE BCC study was based on data up to November 26, 2010.[5] Here we report a 6-month update (non-prespecified; data cut-off May 26, 2011) of the investigator-assessed efficacy and safety endpoints.

METHODS

ERIVANCE BCC study design

- Multicenter, international, nonrandomized, two-cohort study.
- A control group was not used, given the small patient population, historical absence of spontaneous responses, and lack of available effective therapies.
- Patients received oral vismodegib150 mg once daily until disease progression, intolerable toxicity, or study discontinuation.
 - Dose interruption for up to 4 weeks was allowed for the management of toxicity.

Patient eligibility

- At least 18 years of age, with adequate organ function and Eastern Cooperative Oncology Group performance status of ≤2.
- mBCC patients had Response Evaluation Criteria In Solid Tumors (RECIST)-measurable disease (including nodal metastases) using computerized tomography or magnetic resonance imaging.
- laBCC patients had at least one lesion with a longest diameter ≥10 mm that was considered inoperable or for which surgery was considered inappropriate.
 - Surgery was deemed inappropriate if BCC had recurred after ≥2 surgical procedures and curative resection was deemed unlikely, and/or there was substantial morbidity and/or deformity anticipated from surgery.
- In the laBCC cohort, prior radiotherapy to ≥1 target lesion was required, unless contraindicated or inappropriate.

Assessments

- Physical examinations were performed on all patients at baseline and every 4 weeks thereafter.
- For laBCC, a novel composite endpoint for response rate was devised to evaluate therapeutic response; response was defined as meeting any of the following criteria: ≥30% reduction in tumor size by physical examination and/or radiography, and/or complete resolution of ulceration present at baseline.
- In patients with mBCC or radiologically evaluable laBCC, responses were assessed using RECIST (Version 1.0)[6] criteria.
- Efficacy and safety analyses have been performed using data from two cut-off dates:
 - November 26, 2010: 9 months following the first treatment of the last enrolled patient (primary analysis)
 - May 26, 2011: 15 months following the first treatment of the last enrolled patient (6-month update).
- The efficacy-evaluable population included all treated patients for whom the independent pathologist confirmed BCC in archival tumor tissue or baseline biopsy.
- Adverse-event (AE) data were collected for all patients from the initial treatment with vismodegib until data cut-off on May 26, 2011. AEs were graded according to the National Cancer Institute's Common Terminology Criteria for Adverse Events (Version 3.0).

Endpoints

- Endpoints assessed by the investigator included:
 - ORR
 - Duration of response (DOR)
 - Progression-free survival (PFS)
 - Overall survival (OS).

RESULTS

Patient characteristics

- One hundred four patients were enrolled over 13 months at 31 sites in the USA, Europe, and Australia (n=33 for mBCC cohort; n=71 for laBCC).
- Eight patients with laBCC were excluded from efficacy analysis because the independent pathologist did not identify BCC in biopsies taken at baseline, or in archival tissue or post-baseline biopsy. No patients with mBCC were excluded. Baseline patient characteristics are shown in Table 1.

Efficacy

- At the 6-month update, investigator-assessed ORRs for the two groups remained very similar to those reported at the primary analysis (Table 2). The only difference was the conversion of one patient with mBCC from stable disease at the primary analysis date to a partial response at the 6-month update.

Table 1. Demographics and baseline characteristics (efficacy-evaluable population)

		mBCC (n=33)	laBCC (n=63)
Age	Mean (SD)	61.6 (11.4)	61.4 (16.9)
	Median	62.0	62.0
	(range)	(38–92)	(21–101)
Sex	Male	24 (72.7)	35 (55.6)
	Female	9 (27.3)	28 (44.4)
Race	White	33 (100)	63 (100)
laBCC	Inoperable	–	24 (38.1)
	Surgery inappropriate	–	39 (61.9)
	Multiple recurrence	–	16 (25.4)
	Significant morbidity/deformity	–	32 (50.8)
	Radiation previously administered	–	13 (20.6)
	Radiation inappropriate/contraindicated	–	50 (79.4)

Values are given as n (%) unless otherwise stated.
BCC, basal cell carcinoma; laBCC, locally advanced BCC; mBCC, metastatic BCC; SD, standard deviation.

Table 2. Efficacy endpoints with vismodegib in advanced BCC, by cut-off date (efficacy-evaluable population)

	mBCC (n=33)		laBCC (n=63)	
	Nov 26, 2010	May 26, 2011	Nov 26, 2010	May 26, 2011
ORR, n (%) [95% CI]	15 (45.5) [28.1–62.2]	16 (48.5) [30.8–66.2]	38 (60.3) [47.2–71.7]	38 (60.3) [47.2–71.7]
Complete response, n	0	0	20	20
Partial response, n	15	16	18	18
Stable disease, n	15	14	15	15
Progressive disease, n	2	2	6	6
Median DOR, months [95% CI]	(n=15) 12.9 [5.55–12.91]	(n=16) 12.9 [5.55–NE]	(n=38) 7.6 [7.43–NE]	(n=38) NE [7.62–NE]
Median PFS, months [95% CI]	9.2 [7.39–NE]	9.3 [7.39–16.59]	11.3 [9.46–16.82]	12.9 [10.22–NE]
Median OS, months [95% CI]	NE [13.86–NE]	NE [18.10–NE]	NE [17.61–NE]	NE [NE–NE]

BCC, basal cell carcinoma; CI, confidence interval; DOR, duration of response; laBCC, locally advanced BCC; mBCC, metastatic BCC; NE, non-estimable; ORR, objective response rate; OS, overall survival; PFS, progression-free survival.

- Median DOR in patients with mBCC was similar for both cut-off dates (Table 2). For laBCC patients, median DOR was 7.6 months at the protocol-defined data cut-off (November 26, 2010), although with further follow-up at the 6-month update the median DOR became non-estimable due to censoring as a result of a large proportion of patients who continue to be responders at the time of data cut.
 - Kaplan–Meier estimates for DOR are shown in Figure 1A.
- At the 6-month update, median PFS was 9.3 months (95% confidence interval [CI]: 7.39–16.59 months) for the mBCC cohort, which was similar to that of the primary analysis (Table 2). Among laBCC patients the median PFS had increased by 1.6 months to 12.9 months (95% CI: 10.22 months–non-estimable) at the 6-month update (Table 2).
 - Kaplan–Meier estimates for PFS are shown in Figure 1B.
- With median follow-up times for OS of 16.9 months (95% CI: 15.67–17.71 months) for mBCC and 17.4 months (95% CI: 15.51–18.43 months) for laBCC, the OS data were still immature and median OS was not reached for either patient cohort as of May 26, 2011.
- One-year survival rates were 77.3% (95% CI: 62.48–92.09%) for mBCC patients and 93.1% (95% CI: 86.49–99.63%) for laBCC patients, which were similar to those of the primary analysis.

Safety and tolerability

- As of May 26, 2011, approximately two-thirds of the patients had discontinued from the study period (defined as the period during which patients underwent protocol-specified assessments) and entered the survival follow-up (Table 3).
- Consistent with the primary analysis, at the 6 month follow-up the most frequent reasons for treatment discontinuation were patient decision and adverse events in patients with laBCC and disease progression in the mBCC cohort (Table 3).

Figure 1. Kaplan–Meier plots for investigator-assessed endpoints (efficacy-evaluable population)

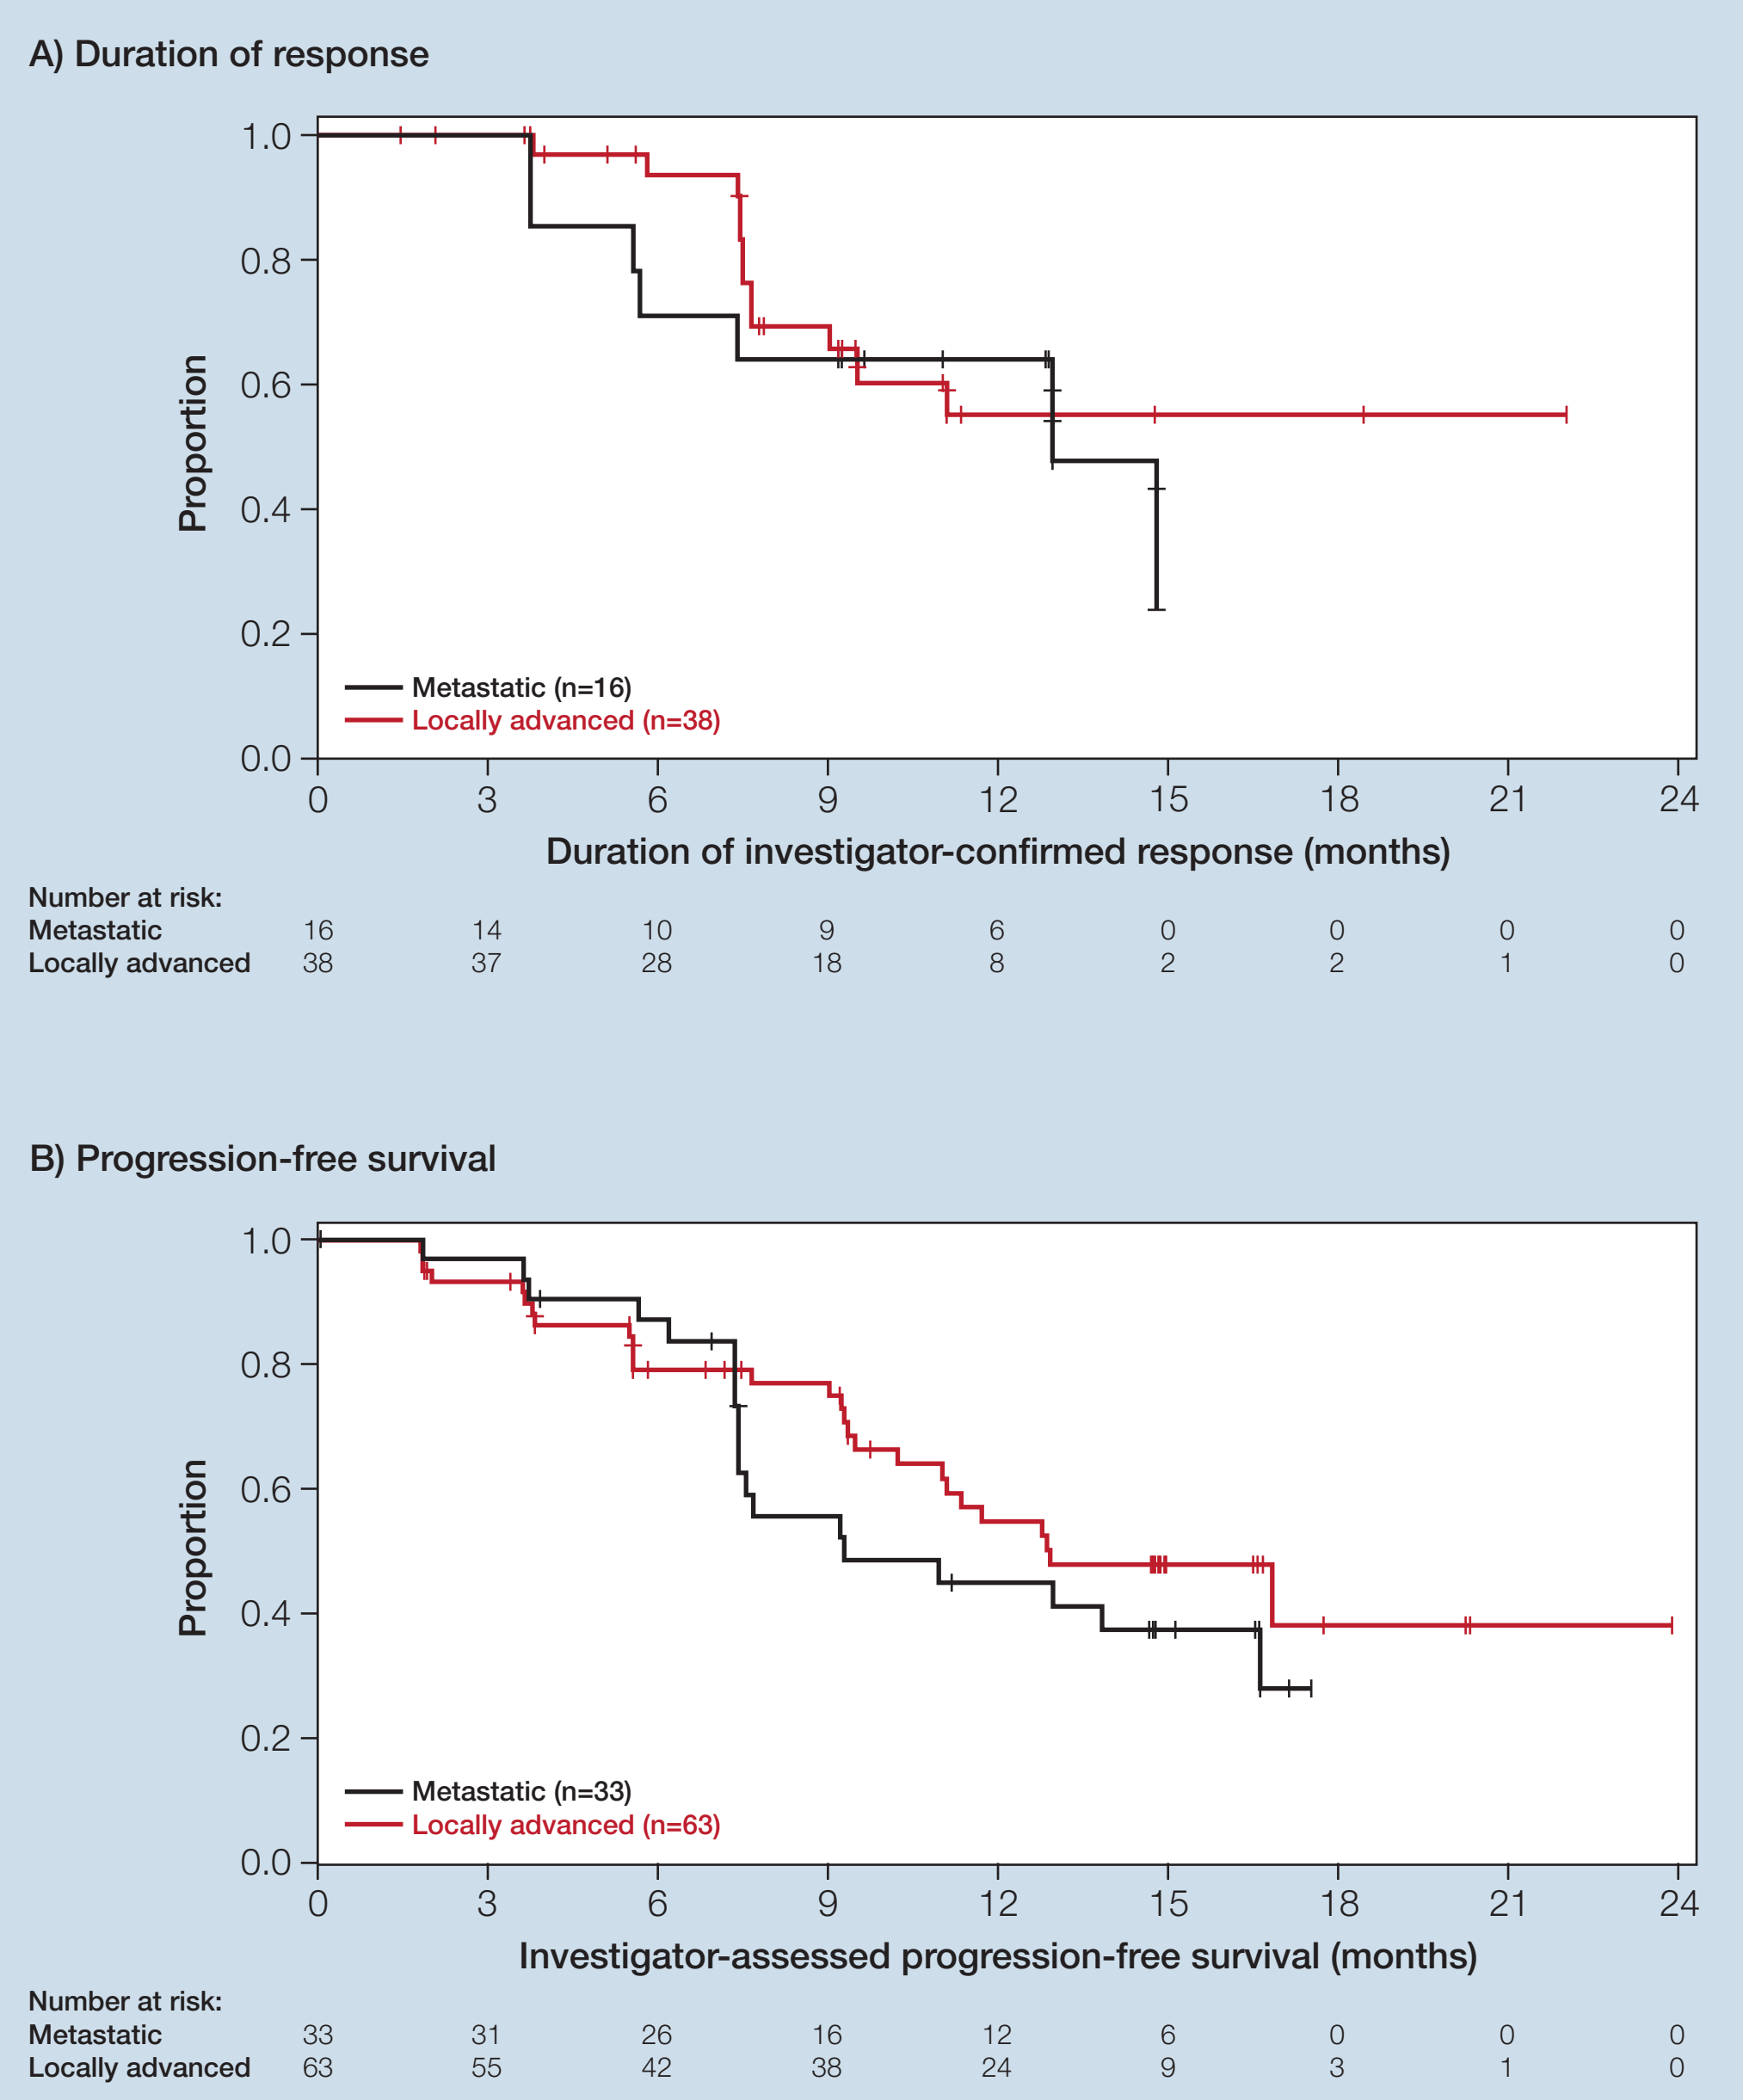
A) Duration of response
Proportion
Metastatic (n=16)
Locally advanced (n=38)
Duration of investigator-confirmed response (months)
Number at risk:
Metastatic 16 14 10 9 6 0 0 0 0
Locally advanced 38 37 28 18 8 2 2 1 0
B) Progression-free survival
Proportion
Metastatic (n=33)
Locally advanced (n=63)
Investigator-assessed progression-free survival (months)
Number at risk:
Metastatic 33 31 26 16 12 6 0 0 0
Locally advanced 63 55 42 38 24 9 3 1 0

Table 3. Reasons for vismodegib discontinuation (all treated patients)

	mBCC (n=33)	laBCC (n=63)
Patients remaining on treatment[a], n (%)	11 (33.3)	26 (36.6)
Total vismodegib treatment discontinuations, n (%)	22 (66.7)	45 (63.4)
Adverse event	2 (6.1)	14 (19.7)
Death	1 (3.0)	2 (2.8)
Lost to follow-up	2 (6.1)	1 (1.4)
Physician decision to discontinue therapy	2 (6.1)	1 (1.4)
Patient decision to discontinue therapy	3 (9.1)	19 (26.8)
Disease progression	12 (36.4)	7 (9.9)
Other	0	1 (1.4)

[a]As of data cut-off: May 26, 2011, 15 months after last patient enrolled. One patient discontinued treatment after the May 26, 2011, data cut-off and is included here as remaining on treatment.
BCC, basal cell carcinoma; laBCC, locally advanced BCC; mBCC, metastatic BCC.

- The median cumulative vismodegib dose received by treated patients increased from 43.2 to 57.6 g between November 26, 2010, and May 26, 2011, and the median duration of drug exposure increased from 9.84 to 12.93 months.
- The vismodegib AE profile for this safety update was consistent with that previously reported (Table 4).

Table 4. Summary of treatment-emergent adverse events with vismodegib, by cut-off date (safety population)

	Cut-off date	
	Nov 26, 2010 (n=104)	May 26, 2011 (n=104)
Treatment-emergent adverse events, n (%)		
Any	104 (100.0)	104 (100.0)
Serious	26 (25.0)	32 (30.8)
Grade ≥3	44 (42.3)	50 (48.1)
Grade 5	7 (6.7)	7 (6.7)
Discontinuation because of treatment-emergent adverse events, n (%)	13 (12.5)	17[a] (16.3)
All deaths, n (%)	16 (15.4)	21 (20.2)

[a]One patient discontinued treatment after the May 26, 2011, data cut-off.

- About 50% of treatment-emergent AEs (TEAEs) were mild to moderate (grade 1/2) in severity (Table 4); the most frequently reported TEAEs did not change between cut-off dates and included muscle spasms (70.2%), alopecia (64.4%), dysgeusia (52.9%), weight decreased (50.0%), fatigue (38.5%), and nausea (32.7%). Since the November 26, 2010, data cut-off, only nasopharyngitis increased in incidence to ≥10% of all patients (from 9.6% [10 patients] to 11.5% [12 patients]).
- The grade 3–5 vismodegib AE profile for this 6-month update was generally consistent with that previously reported (Table 4).
 - No additional grade 5 AEs were reported in this update compared with the primary analysis.
 - In this update 50 patients (48.1%) had reported grade 3–5 AEs compared with 44 patients (42.3%) as of the November 26, 2010, data cut-off.
 - The most common grade 3–5 AEs in this update included decrease in weight (5.8%), muscle spasms (5.8%), and fatigue (4.8%).
- As of the May 26, 2011, data cut-off, a total of 21 patients had died (20.2%) compared with 16 (15.4%) at the November 26, 2010, follow-up (Table 4). All five additional deaths occurred during survival follow-up, with none occurring while the patient was taking study drug or within 30 days of the last dose of study drug.
 - Four of the additional deaths were due to progressive disease, one in a patient with laBCC and three in patients with mBCC. The fifth death resulted from melanoma in a patient in the laBCC group, who had a history of prior melanoma.

CONCLUSIONS

- This 6-month update of investigator-assessed efficacy and safety endpoints from the ERIVANCE BCC study supports the significant clinical benefit of vismodegib in both mBCC and laBCC reported at the primary analysis.
- Vismodegib given at the recommended dose of 150 mg once daily has demonstrated a manageable safety profile in patients with mBCC and laBCC.
- Targeted inhibition of Hedgehog signaling with vismodegib represents a new therapeutic option for patients with advanced BCC.
- Based on the results of the ERIVANCE study, vismodegib has been approved by the US Food and Drug Administration for the treatment of adults with mBCC, or with laBCC that has recurred following surgery or who are not candidates for surgery, and who are not candidates for radiation.

REFERENCES

1. Rogers H. The Skin Cancer Foundation. 2012.
2. Göppner D, Leverkus M. J Skin Cancer 2011:650258.
3. Daya-Grosjean I, Couve-Privat S. Cancer Lett 2005;225:181–192.
4. LoRusso PM, Rudin CM, Reddy JC, et al. Clin Cancer Res 2011;17:2502–2511.
5. Sekulic A, Migden MR, Oro AE, et al. N Engl J Med [In press].
6. Therasse P, Arbuck SG, Eisenhauser EA, et al. J Natl Cancer Inst 2000;92:205–216.

ACKNOWLEDGMENTS

The authors would like to thank the participating investigators, patients, and current and past members of Roche and Genentech vismodegib teams. This study was funded by Roche–Genentech. Support for medical writing assistance was provided by F. Hoffmann–La Roche. Roche is developing vismodegib under a collaboration agreement with Curis, Inc. Vismodegib was discovered by Genentech and jointly validated by Genentech and Curis through a series of preclinical studies. Through this collaboration, Genentech (United States), Roche (ex-United States excluding Japan and Korea), and Chugai Pharmaceuticals (Japan) are responsible for the clinical development and commercialization of vismodegib.

A pdf of this poster can be downloaded using the QR code above

48th Annual ASCO Meeting, Chicago, June 1–5, 2012